Safety and Activity of IPI-504 (retaspimycin hydrochloride) and Docetaxel in Pretreated Patients with Metastatic Non-Small Cell Lung Cancer (NSCLC)

G. J. Riely[1], S. N. Gettinger[2], R. G. Stoller[3], N. Y. Gabrail[4], G. K. Dy[5], G. J. Weiss[6], C. Tunkey[7], G. Skliris[7], S. Strychor[8], J. Dunbar[7], D. DeLucia[7], R. W. Ross[7], J. E. Gray[9]

[1]Memorial Sloan-Kettering Cancer Center, New York, NY; [2]Yale University School of Medicine, New Haven, CT; [3]University of Pittsburgh Cancer Center, Pittsburgh, PA; [4]Gabrail Cancer Center, Canton, OH; [5]Roswell Park Cancer Institute, Buffalo, NY; [6]Virginia G. Piper Cancer Center at Scottsdale Healthcare/TGen, Scottsdale, AZ; [7]Infinity Pharmaceuticals, Inc., Cambridge, MA; [8]University of Pittsburgh, Pittsburgh, PA; [9]H. Lee Moffitt Cancer Center & Research Institute, Tampa, FL

Background

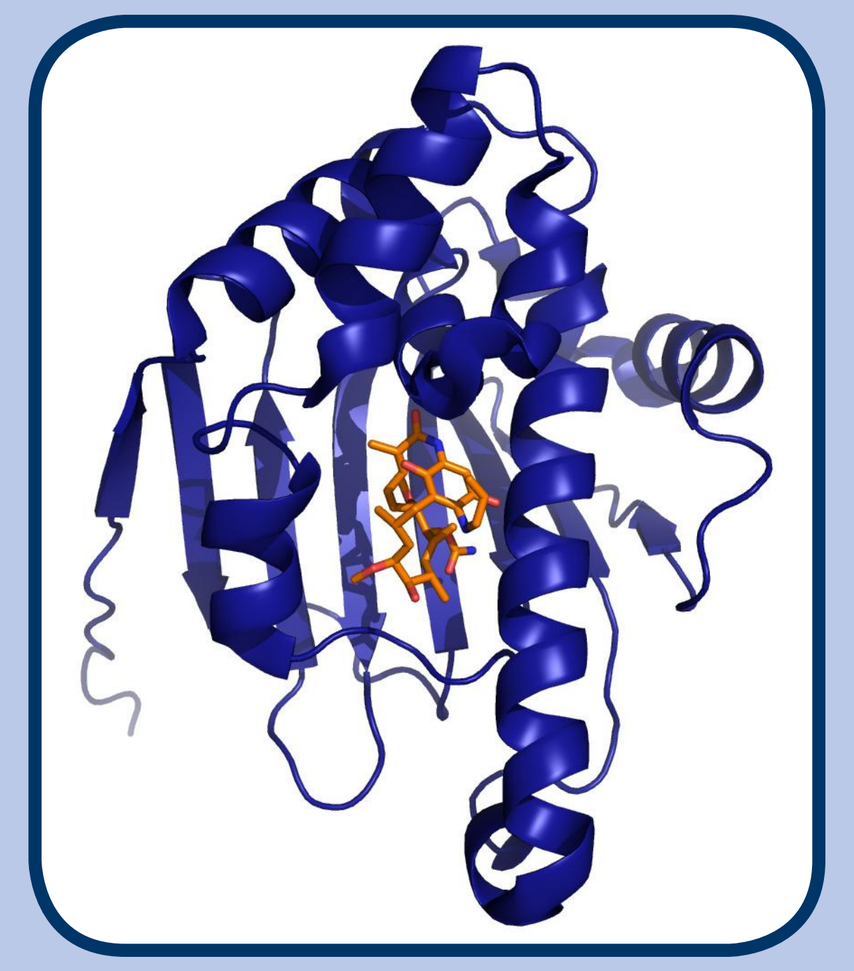

- Hsp90
 - Hsp90 is a chaperone protein necessary for stability and function of certain client proteins.
 - Many oncoproteins are dependent on Hsp90 for function.
- Retaspimycin HCl (IPI-504)
 - Water-soluble analog of the natural product geldanamycin.
 - Single-agent response rate of 7% in patients with metastatic NSCLC (N=76). In 40 patients with EGFR wild-type NSCLC, the response rate was 10%. Among 3 patients with known ALK rearrangements, 2 had a partial response to retaspimycin HCl as a single agent and a third patient had stable disease (24% reduction) durable for 7 months. (Sequist et al, J Clin Oncol 28:4953-60)
- Therapeutic Rationale
 - A constitutively activated heat shock response is an essential property of cancer cells (vs. normal cells) and is necessary for their growth, proliferation, and survival. Hsp90 is an essential component of that heat shock response.
 - Certain anticancer therapies may enhance the dependency of cancer cells on the Hsp90 chaperone.
 - Docetaxel is an approved agent for relapsed NSCLC, with a response rate of ~ 8% and a PFS of ~ 3 months. (Hanna et al, J Clin Oncol, 22:1589-97)
 - Combining an Hsp90 inhibitor such as retaspimycin HCl with cytotoxic chemotherapy may result in enhanced cancer cell-specific killing. Results from multiple pre-clinical models support the clinical evaluation of the combination of Hsp90 inhibitors and taxanes. (Sawai et al, Cancer Res 68:589-96; Sain et al, Mol Cancer Ther 5:1197-208; Nguyen et al, Ann Thorac Surg 72:371-8)

Study Design

- Objectives
 - Determine the safety and anti-tumor activity of retaspimycin HCl + docetaxel in patients with pretreated NSCLC
- Study Design
 - An expansion of a Phase 1b trial of retaspimycin HCl in combination with docetaxel in patients with pretreated NSCLC (Riely, ASCO 2009, Abst #3547)
- Key Eligibility Criteria
 - Adults with pretreated metastatic NSCLC and no prior exposure to docetaxel
 - Progressive disease; clinically stable brain metastases allowed
 - Measurable disease by RECIST criteria
 - Karnofsky performance status ≥ 70
- Dosing Schedule
 - Retaspimycin HCl: 300 mg/m² IV QW
 - Docetaxel: 75 mg/m² IV Q3W
 - Growth factor support was allowed after Cycle 1

Baseline Demographics

Total Enrolled, n	23
Age, years†	61 (31 - 77)
Women, n (%)	13 (57)
Time Since Initial Diagnosis of NSCLC, months†	11 (2 - 32)
Smoking Status, n (%)	
Never	5 (22)
Former or Current	18 (78)
Tobacco Exposure, pack-years†	30 (15 - 56)
NSCLC Histological Subtype, n (%)	
Adenocarcinoma	13 (57)
Squamous	7 (30)
Unspecified	2 (9)
Large Cell	1 (4)
Number of Prior Lines of Therapy, n (%)	
1	12 (52)
2	4 (17)
3	7 (30)
Number of Prior Chemotherapies†	3 (1 - 7)
EGFR Sequencing, n (%)	5 (22)
EGFR Wild-Type	5 (100)
KRAS Sequencing, n (%)	17 (74)
KRAS Wild-Type	11 (65)

† Median (range)

Pharmacokinetics

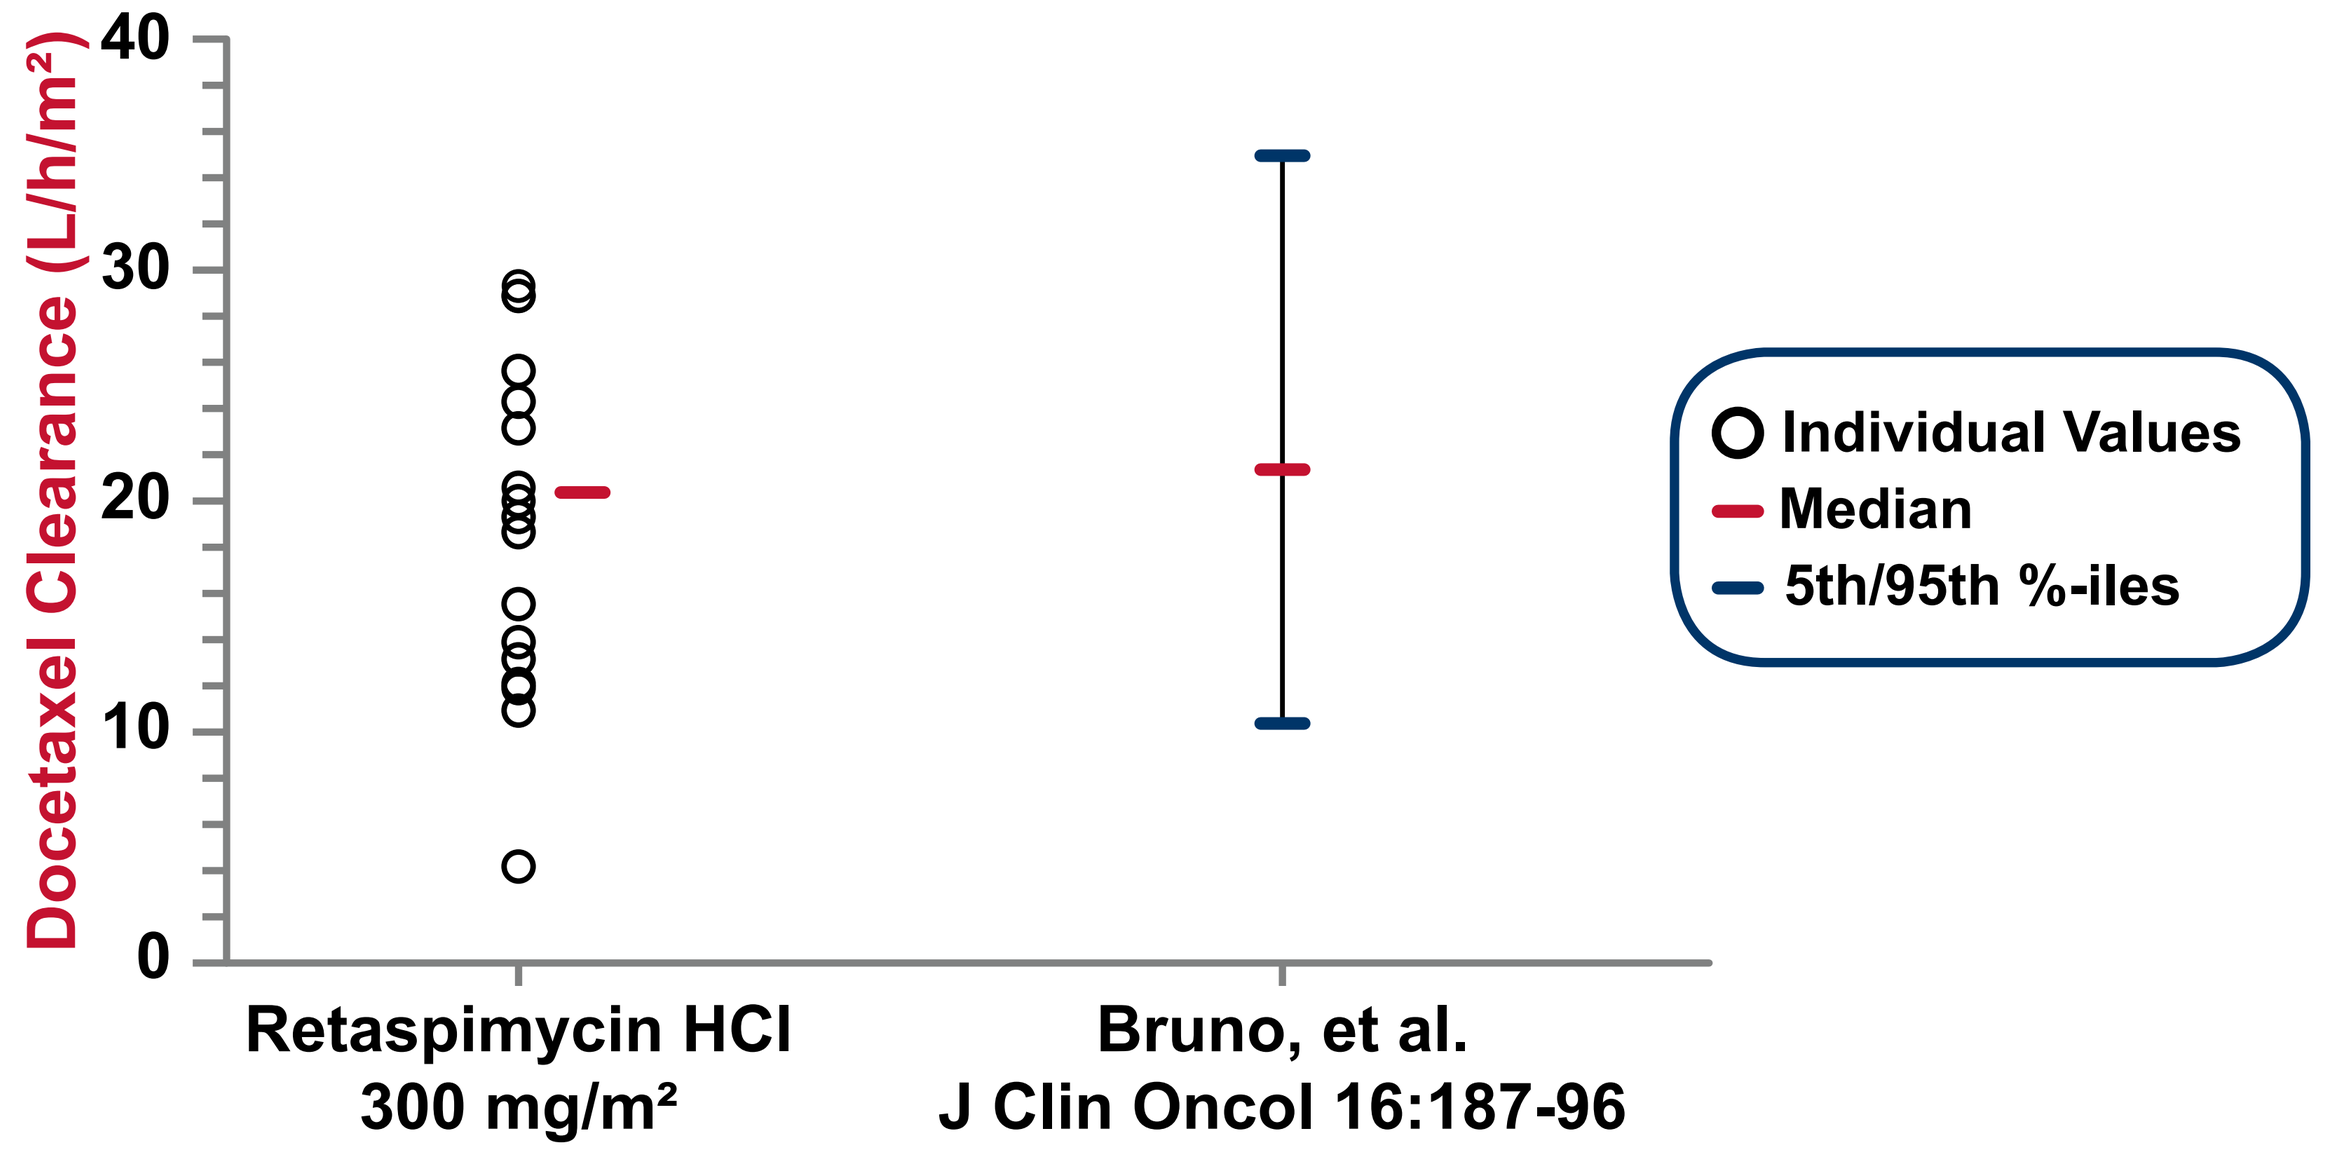
Docetaxel Clearance (L/h/m²)
40
30
20
10
0
Retaspimycin HCl 300 mg/m²
Bruno, et al. J Clin Oncol 16:187-96
Individual Values
Median
5th/95th %-iles

- Docetaxel clearance was unaltered by retaspimycin HCl co-administration.

Safety

Related TEAEs in ≥ 5% of Patients	Worst CTCAE Grade on Study, n (%)		
	1 or 2	3	4
Fatigue	13 (57)	2 (9)	0 (0)
Diarrhea	7 (30)	2 (9)	0 (0)
Nausea	6 (26)	3 (13)	0 (0)
Vomiting	5 (22)	3 (13)	0 (0)
Anemia	7 (30)	1 (4)	0 (0)
Neutropenia	1 (4)	1 (4)	6 (26)
Dysgeusia	5 (22)	0 (0)	0 (0)
Headache	5 (22)	0 (0)	0 (0)
Alopecia	5 (22)	0 (0)	0 (0)
Anorexia	5 (22)	0 (0)	0 (0)
Mucosal inflammation	4 (17)	0 (0)	0 (0)
Arthralgia	3 (13)	1 (4)	0 (0)
Constipation	3 (13)	0 (0)	0 (0)
Edema peripheral	3 (13)	0 (0)	0 (0)
Dizziness	3 (13)	0 (0)	0 (0)
Muscular weakness	2 (9)	1 (4)	0 (0)
Myalgia	3 (13)	0 (0)	0 (0)
Epistaxis	3 (13)	0 (0)	0 (0)

TEAE = treatment-emergent adverse event
CTCAE = Common Terminology Criteria for Adverse Events

- 23 patients received a median of 2 treatment cycles (range: 1 - 14).
- No patient had a treatment-related Grade 3 or 4 AST, ALT, or bilirubin on study.
- No drug-related deaths or discontinuations occurred.

Patient Disposition

Patients Treated, n	23
Patients On Study, n (%)	2 (9)
Discontinuations, n (%)	21 (91)
Radiographic Disease Progression	14 (61)
Clinical or Symptomatic Progression	2 (9)
Withdrawal by Patient	2 (9)
Physician Decision	2 (9)
Death	1 (4)

Activity

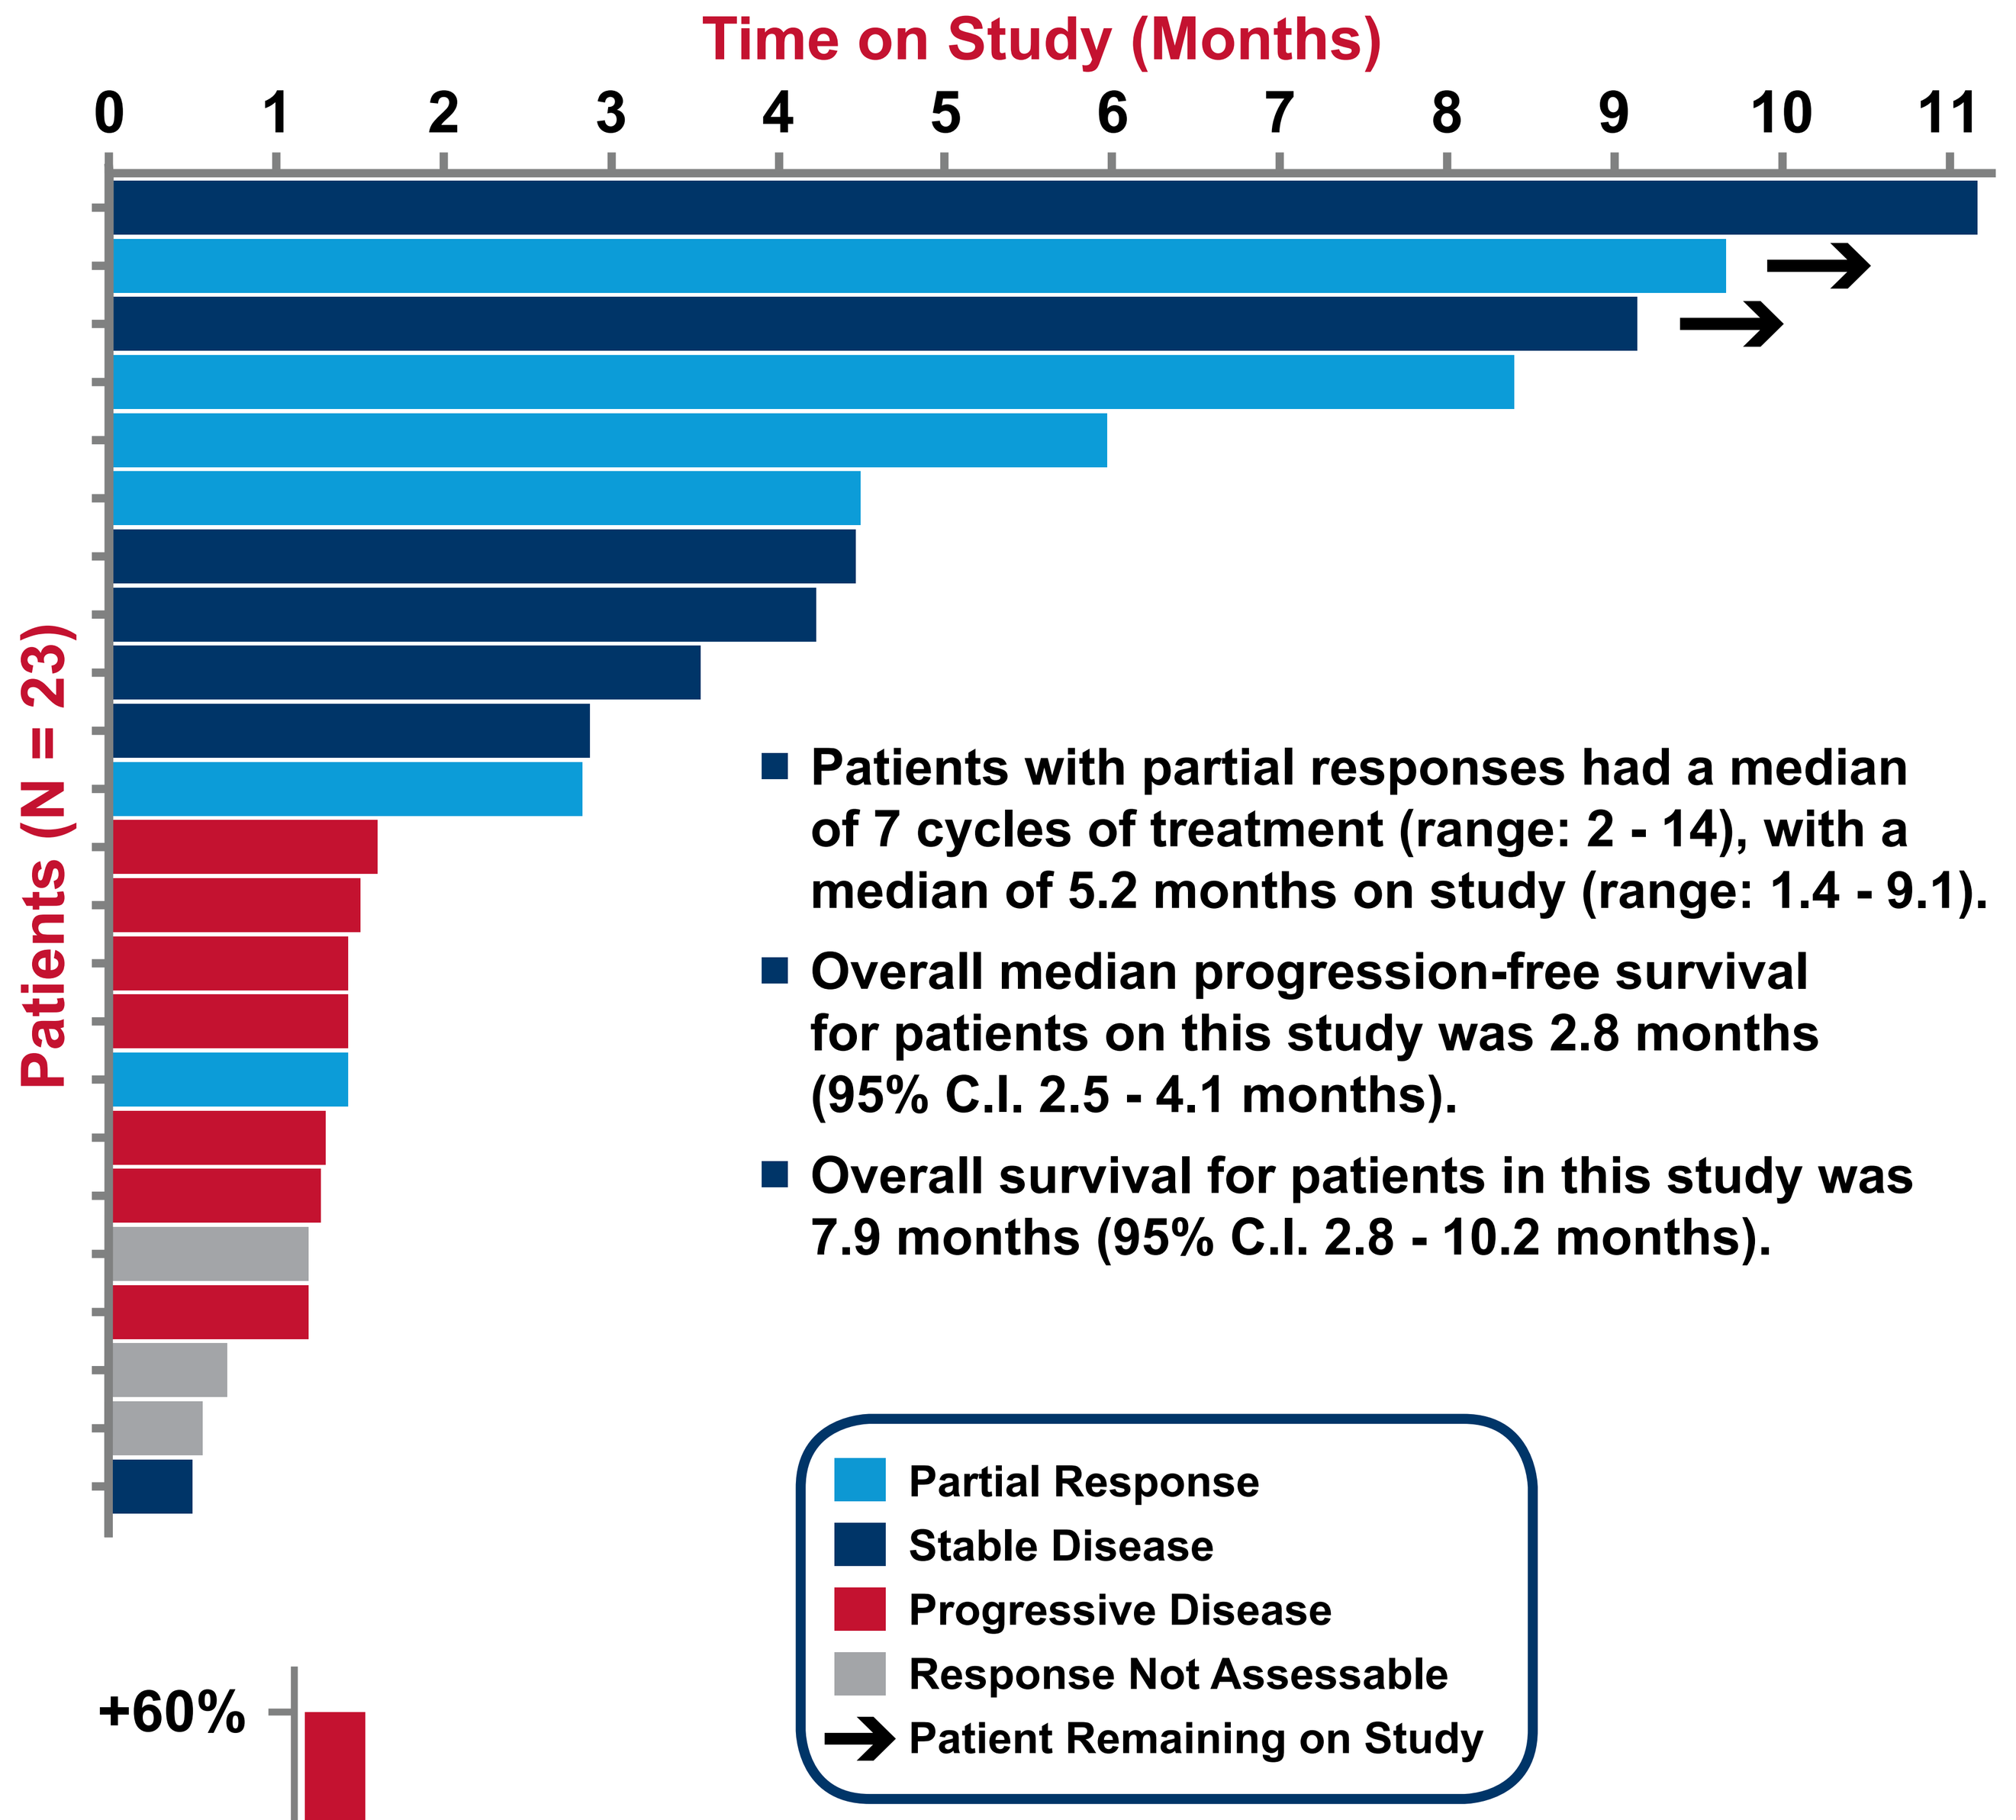
Time on Study (Months)
0 1 2 3 4 5 6 7 8 9 10 11
Patients (N = 23)
Partial Response
Stable Disease
Progressive Disease
Response Not Assessable
Patient Remaining on Study

- Patients with partial responses had a median of 7 cycles of treatment (range: 2 - 14), with a median of 5.2 months on study (range: 1.4 - 9.1).
- Overall median progression-free survival for patients on this study was 2.8 months (95% C.I. 2.5 - 4.1 months).
- Overall survival for patients in this study was 7.9 months (95% C.I. 2.8 - 10.2 months).

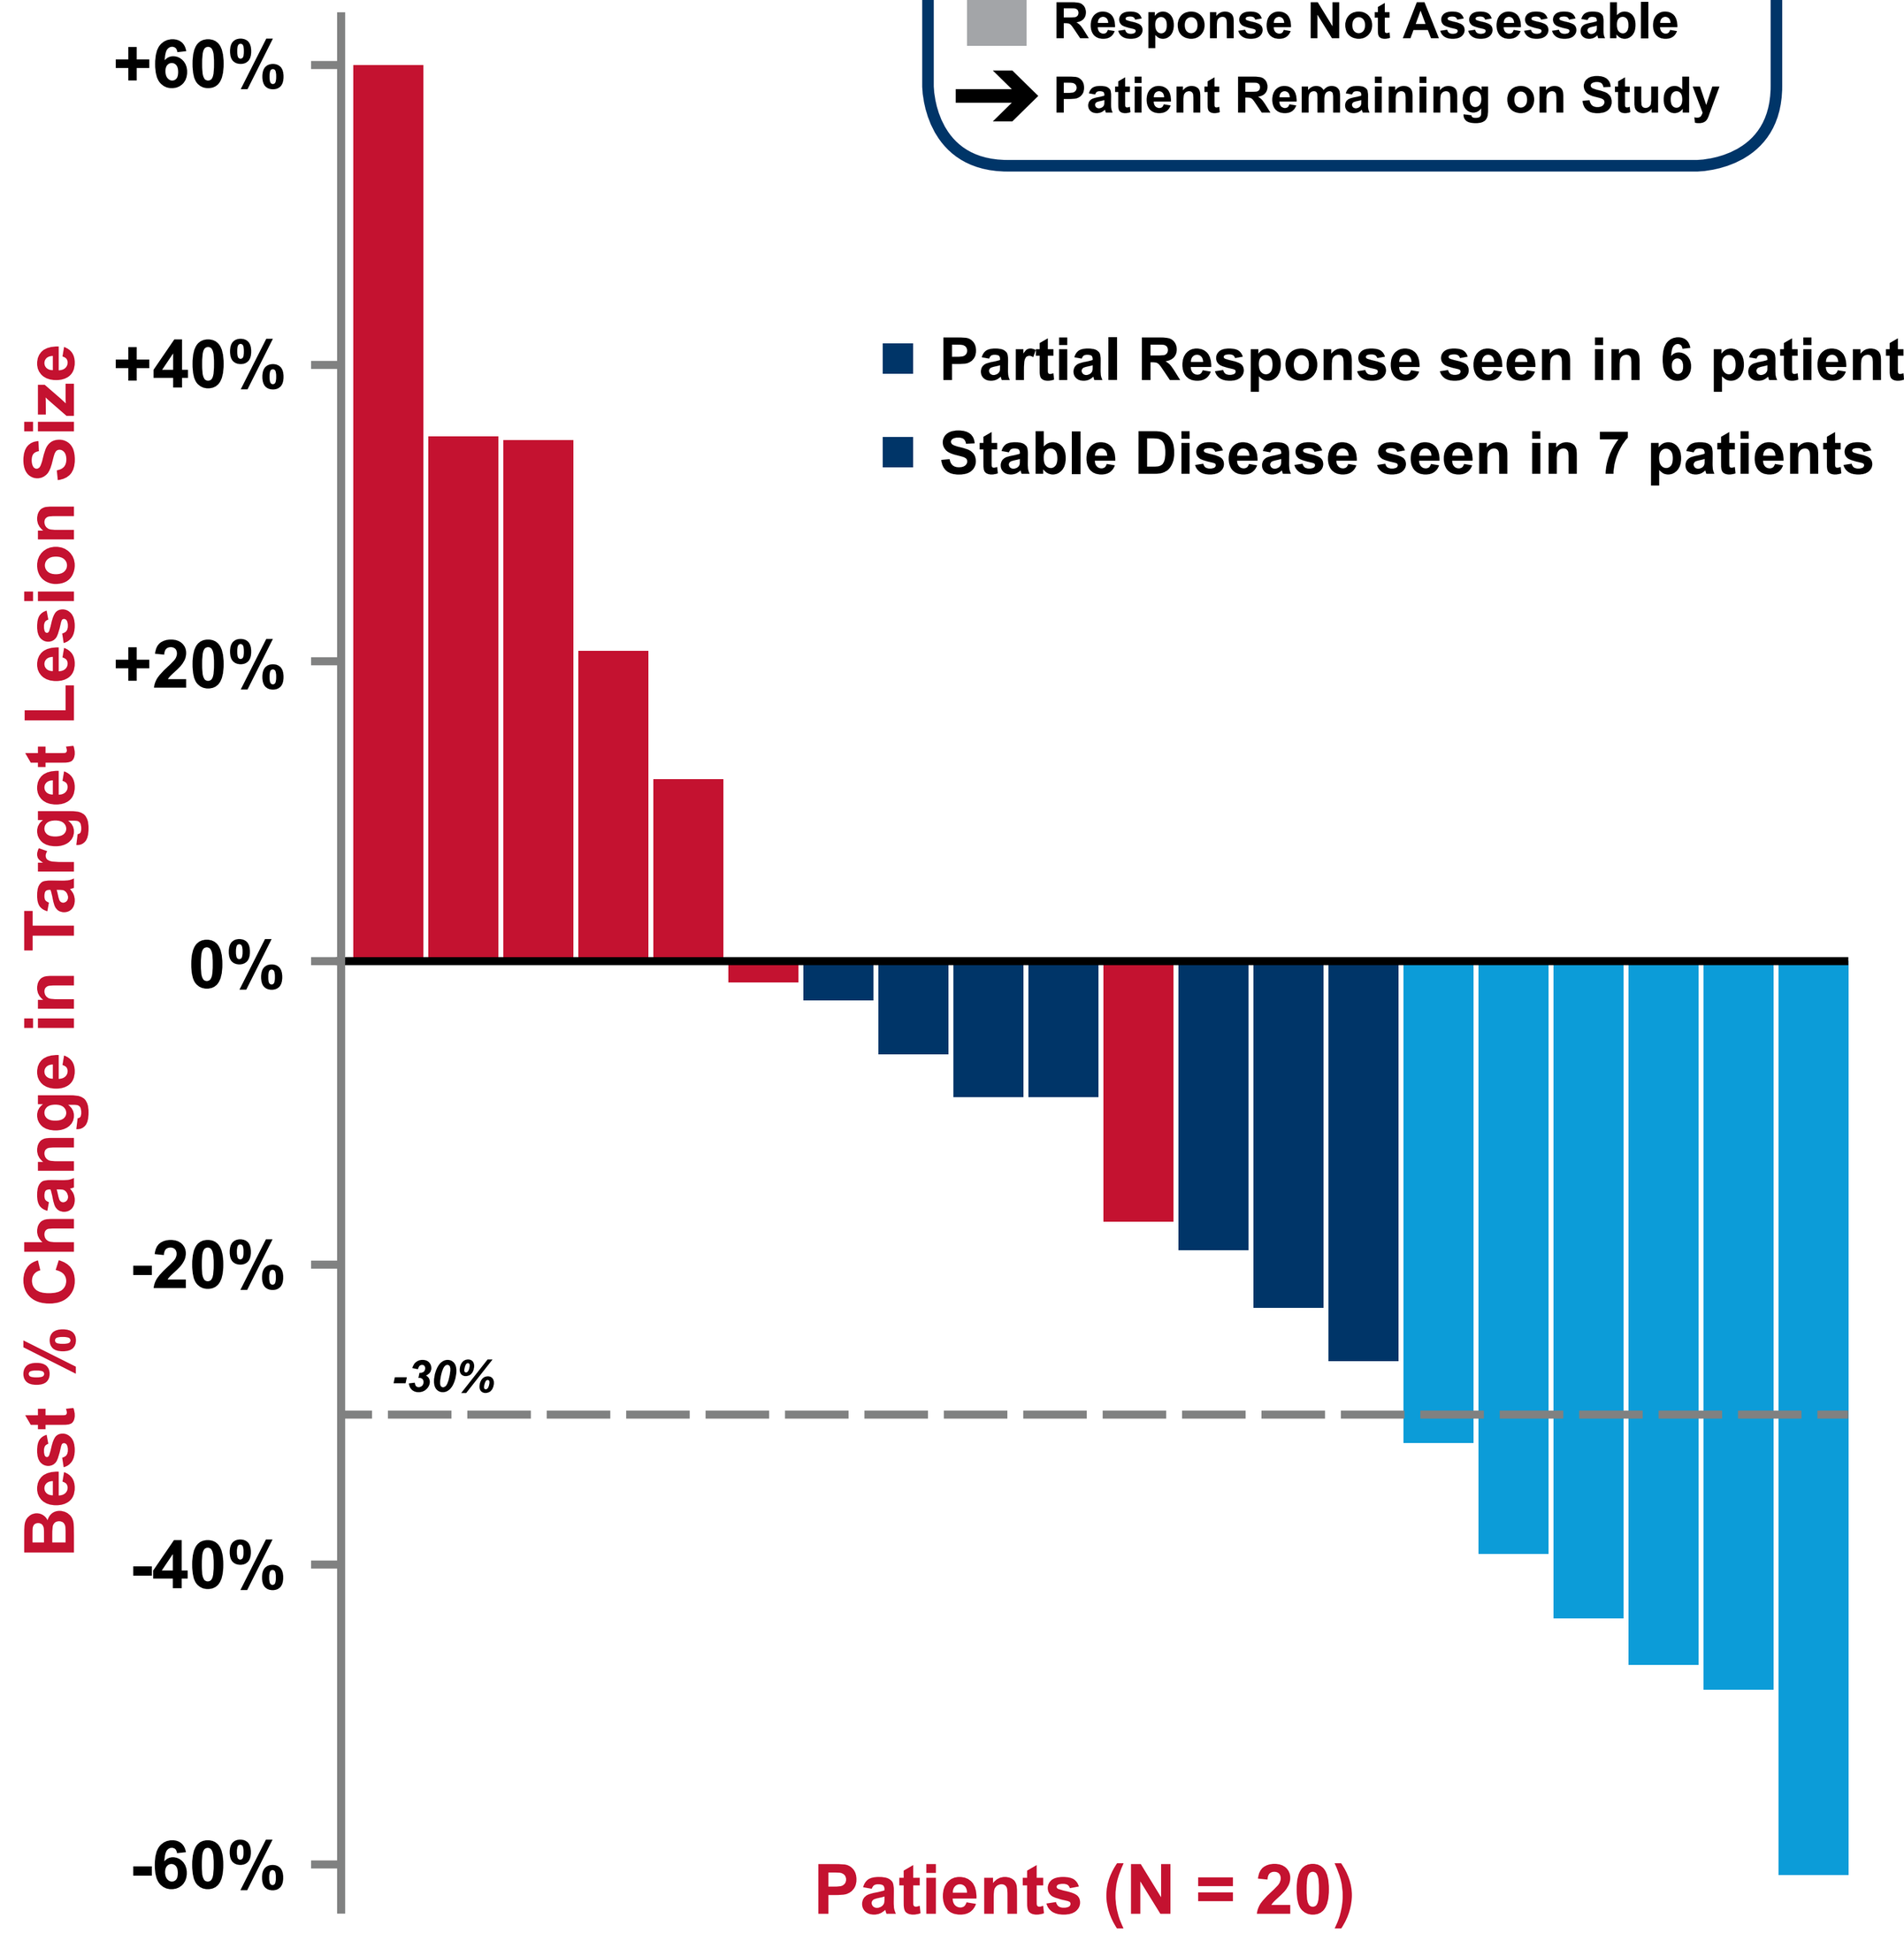
Best % Change in Target Lesion Size
+60%
+40%
+20%
0%
-20%
-40%
-60%
-30%
Patients (N = 20)

- Partial Response seen in 6 patients (ORR = 26%)
- Stable Disease seen in 7 patients

- In exploratory analyses, patients with the highest response rates fell into several overlapping categories:
 - Current or former smokers (ORR = 33%)
 - Those with tumors that were KRAS wild-type (ORR = 36%)
 - Those with squamous cell carcinoma (ORR = 43%)

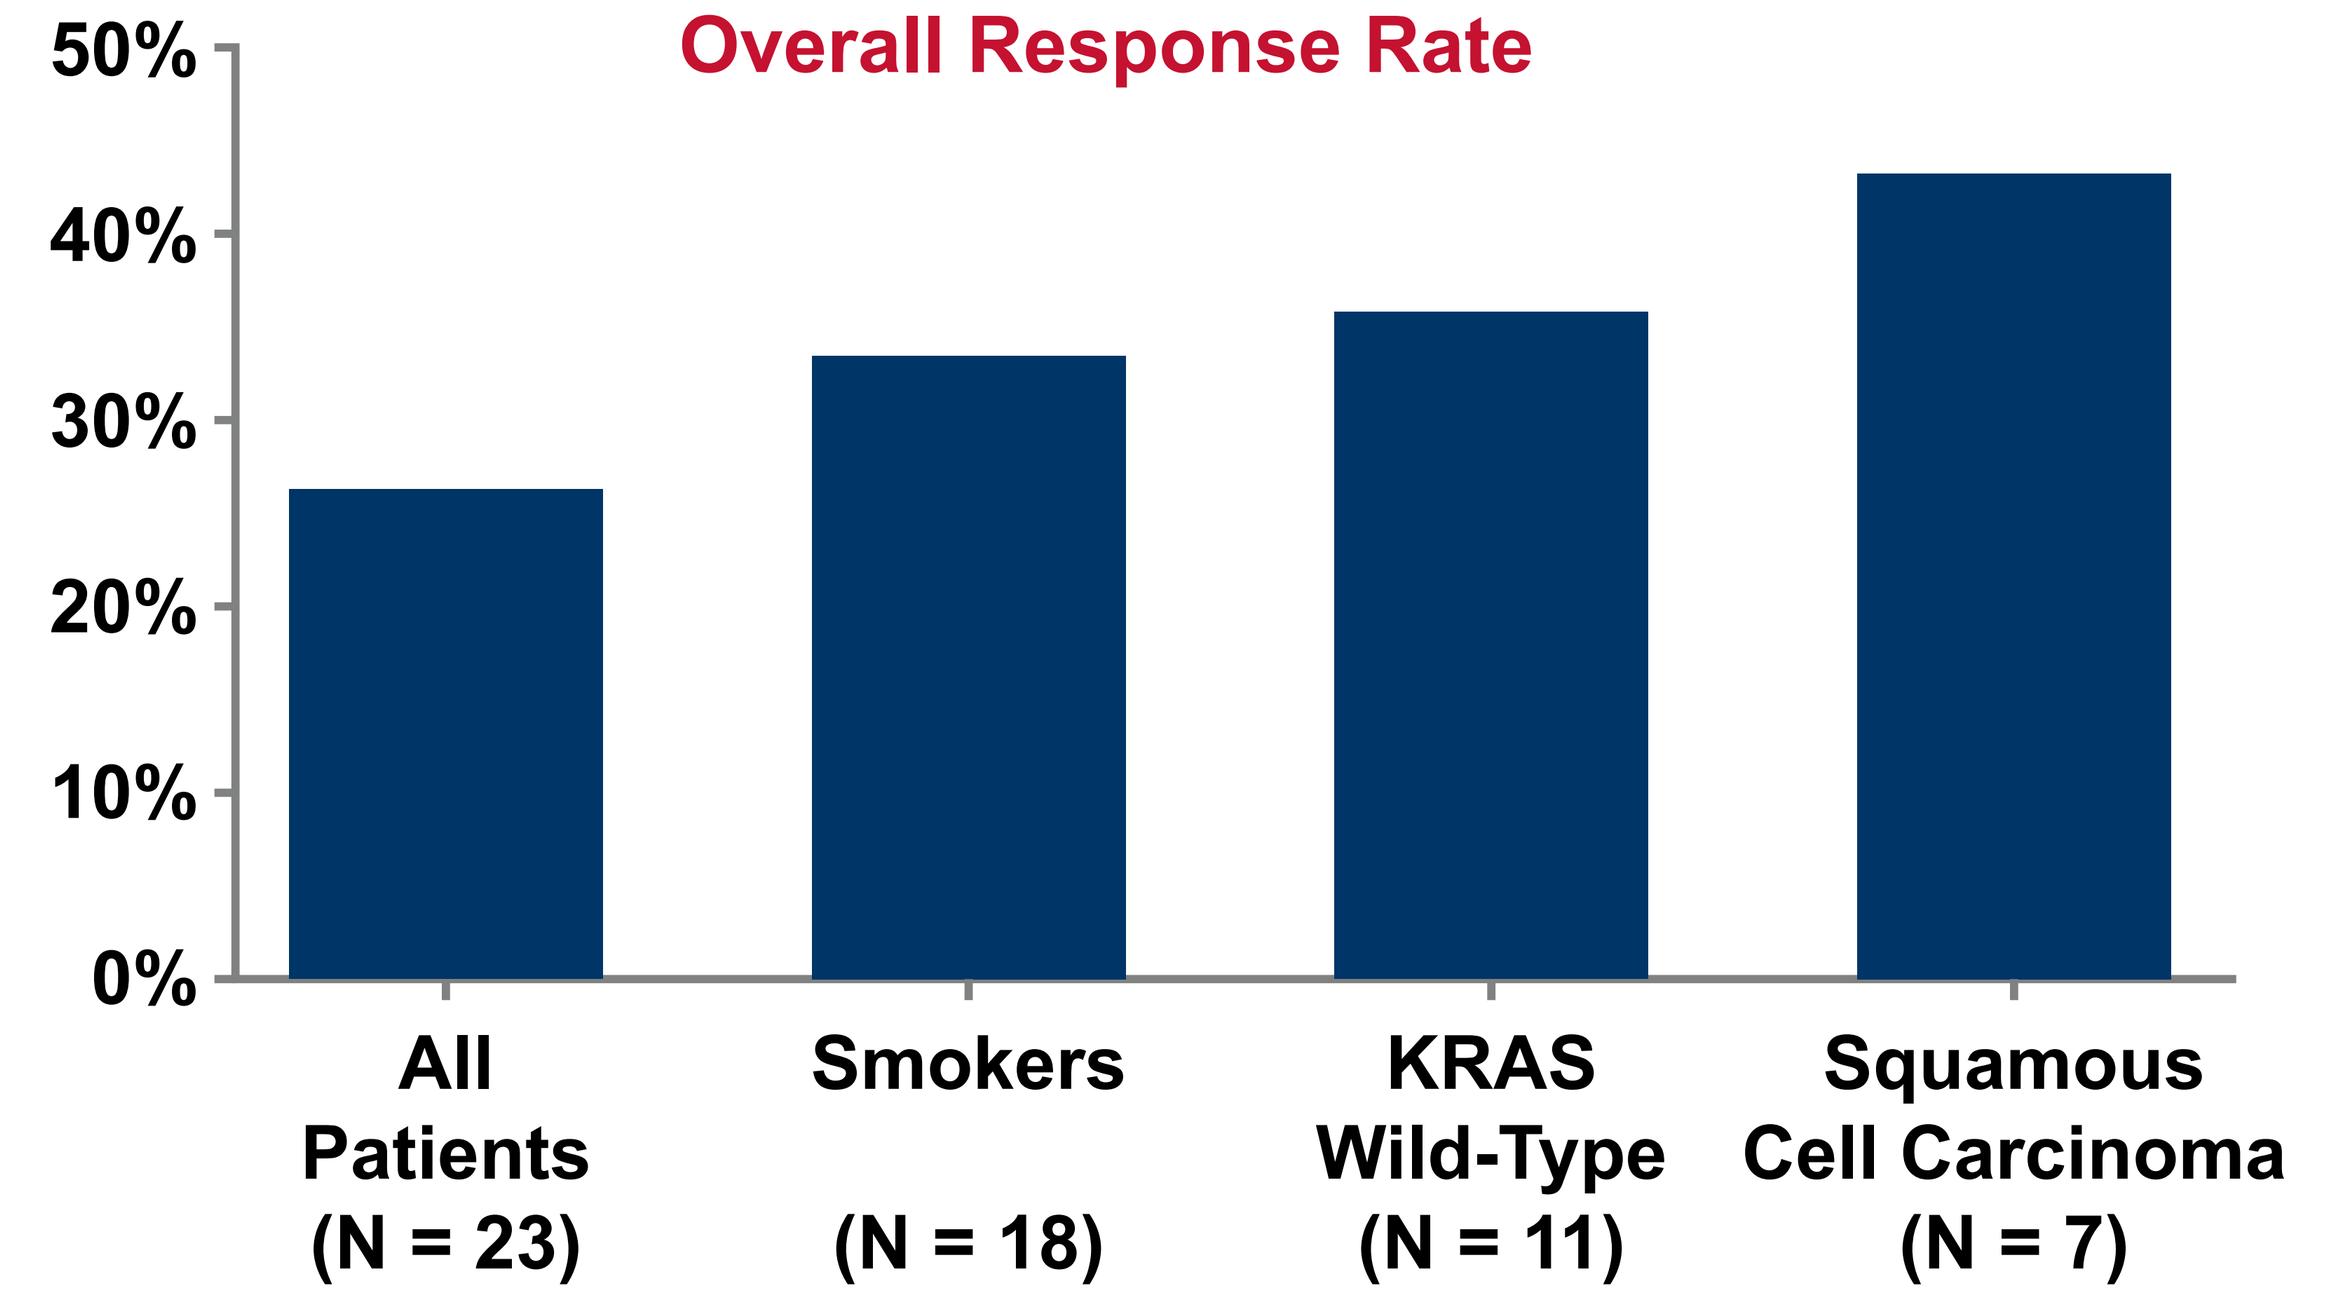
Overall Response Rate
50%
40%
30%
20%
10%
0%
All Patients (N = 23)
Smokers (N = 18)
KRAS Wild-Type (N = 11)
Squamous Cell Carcinoma (N = 7)

Conclusions

- The combination of weekly retaspimycin HCl (300 mg/m²) + docetaxel (75 mg/m² every 3 weeks) was well-tolerated in patients with NSCLC, with no unexpected or overlapping toxicities.
- Retaspimycin HCl given on a weekly schedule was convenient and well-tolerated. In particular:
 - Liver function test increases related to retaspimycin HCl were all mild in nature, and none led to dose reduction or discontinuation from the trial.
 - Gastrointestinal toxicity was manageable and primarily low grade (Grade 1 or 2) and did not lead to study discontinuation.
 - No significant ocular toxicities were identified.
- Clinical activity of the combination was noted in pretreated patients with NSCLC, including:
 - ORR of 26% as compared with a historical docetaxel response rate of approximately 8%.
 - Higher response rates were observed in patients with historically poorer outcomes, such as heavy smokers and those with squamous cell histology.
- Based on these data, an adaptive randomized Phase 2 study is underway comparing retaspimycin HCl + docetaxel versus placebo + docetaxel in pretreated patients with NSCLC and a smoking history. This trial is designed to identify and validate potential predictive biomarkers of the activity of combination treatment.

Poster presented at the American Society of Clinical Oncology (ASCO) Annual Meeting, June 3-7, 2011, Chicago, Illinois